#TIP4

Treatment of Advanced Renal Cell Carcinoma After Progression on Systemic Therapy: Open-Label Phase 2 Study of 2 Doses of the HIF-2α Inhibitor Belzutifan

Michael B. Atkins[1]; Yanfang Liu[2]; Rodolfo Perini[2]; Ananya Roy[2]; John Haanen[3]

[1]Georgetown Lombardi Comprehensive Cancer Center, Washington, DC, USA; [2]Merck & Co., Inc., Kenilworth, NJ, USA; [3]Netherlands Cancer Institute, Amsterdam, Netherlands

Background

- Approximately 90% of clear cell renal cell carcinoma (ccRCC) is caused by a loss of function in the von Hippel-Lindau (*VHL*) gene, leading to the accumulation of hypoxia-inducible factor 2α (HIF-2α) and driving tumor growth in ccRCC (**Figure 1**)[1,2]
 - HIF-2α activates genes associated with invasion and metastasis, cell survival, resistance to the immune system, and angiogenesis[3]
 - Treatment options for patients with advanced ccRCC are limited after immunotherapy and vascular endothelial growth factor (VEGF)–targeted therapy
- Belzutifan (MK-6482) is a small molecule HIF-2α inhibitor that has shown antitumor activity in patients with advanced ccRCC after progression with other systemic therapies in a phase 1/2 study[4]
 - Patients received belzutifan 120 mg orally once daily (QD); objective response rate (ORR) was 25%, and median progression-free survival (PFS) was 14.5 months
 - The median (range) number of prior systemic therapies was 3 (1-9); 34 patients (62%) had previously received ≥3 therapies
 - 44 patients (80%) had received anti–PD-1 therapies and 50 patients (91%) had received anti-VEGF therapies before enrollment
 - 39 patients (71%) received a VEGF/VEGF receptor inhibitor and an immune checkpoint inhibitor
- This randomized, open-label, phase 2 study (NCT04489771) was conducted to evaluate the efficacy and safety of 2 doses of belzutifan in patients with advanced ccRCC who experienced disease progression after systemic therapy

Figure 1. The Role of Belzutifan in Inhibiting the HIF-2α Pathway

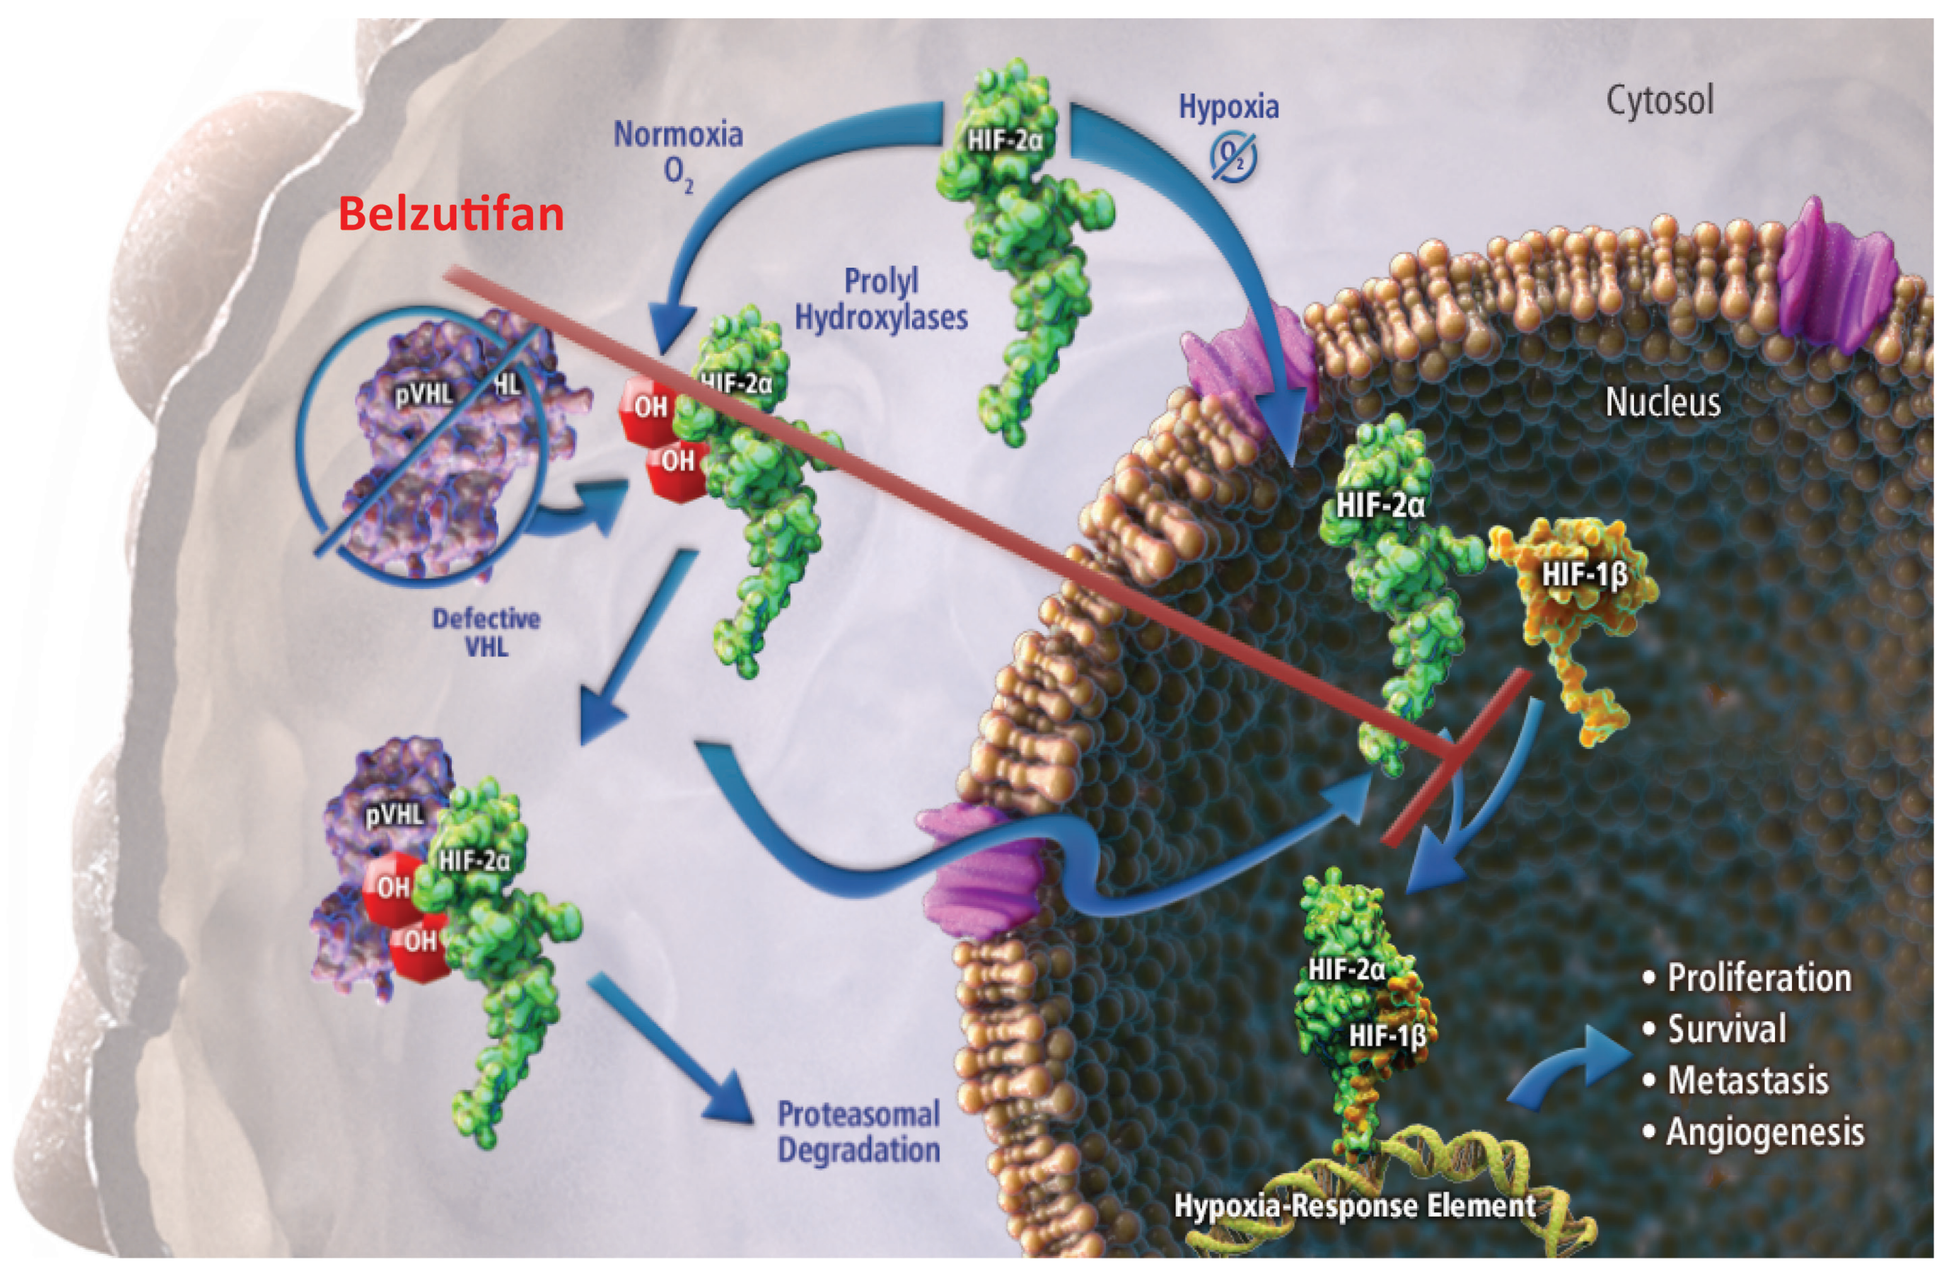
Belzutifan
Normoxia
O2
HIF-2α
Hypoxia
Cytosol
Prolyl Hydroxylases
pVHL
OH
Defective VHL
Nucleus
HIF-1β
Proteasomal Degradation
• Proliferation
• Survival
• Metastasis
• Angiogenesis
Hypoxia-Response Element

pVHL, von Hippel-Lindau tumor suppressor.

Objectives

Primary

- To compare the antitumor activity of the 120-mg QD dose and 200-mg QD dose of belzutifan for the treatment of patients with advanced ccRCC, measured by ORR per RECIST v1.1 by blinded independent central review (BICR)

Secondary

- To compare the following between the 120-mg QD and 200-mg QD doses of belzutifan for the treatment of patients with advanced ccRCC
 - PFS per RECIST v1.1 by BICR
 - Duration of response (DOR) per RECIST v1.1 by BICR
 - Clinical benefit rate (CBR), defined as the percentage of patients who achieve complete response, partial response, or stable disease of ≥6 months, evaluated per RECIST v1.1 by BICR
 - Overall survival (OS)
 - Safety and tolerability
 - Pharmacokinetics (PK)

Exploratory

- To identify molecular (genomic, metabolic, and/or proteomic) biomarkers that could be associated with clinical response/resistance, safety, pharmacodynamic activity, and/or the mechanism of action of belzutifan

Methods

Study Design

- Approximately 150 adults with advanced ccRCC that has progressed after a maximum of 3 prior systemic therapies will be enrolled in this randomized, open-label, multicenter, phase 2 study (**Figure 2**)
 - Prior systemic therapy must include a first-line anti–PD-1/anti–PD-L1 + cytotoxic T-lymphocyte–associated protein 4 (CTLA-4) or anti–PD-1/anti–PD-L1 + VEGF-targeted tyrosine kinase inhibitor (TKI) combination
- Eligible patients will be randomly assigned 1:1 to receive belzutifan 120 mg or 200 mg QD; ~75 patients will be included in each treatment arm
 - At randomization, patients will be stratified by International mRCC Database Consortium (IMDC) prognostic scores (0 vs 1 or 2 vs 3-6) and by number of prior TKI therapies for advanced ccRCC (0 vs 1 vs 2 or 3)
- Study treatment will continue until documented disease progression, unacceptable toxicity, or withdrawal of the patient

Figure 2. Study Design

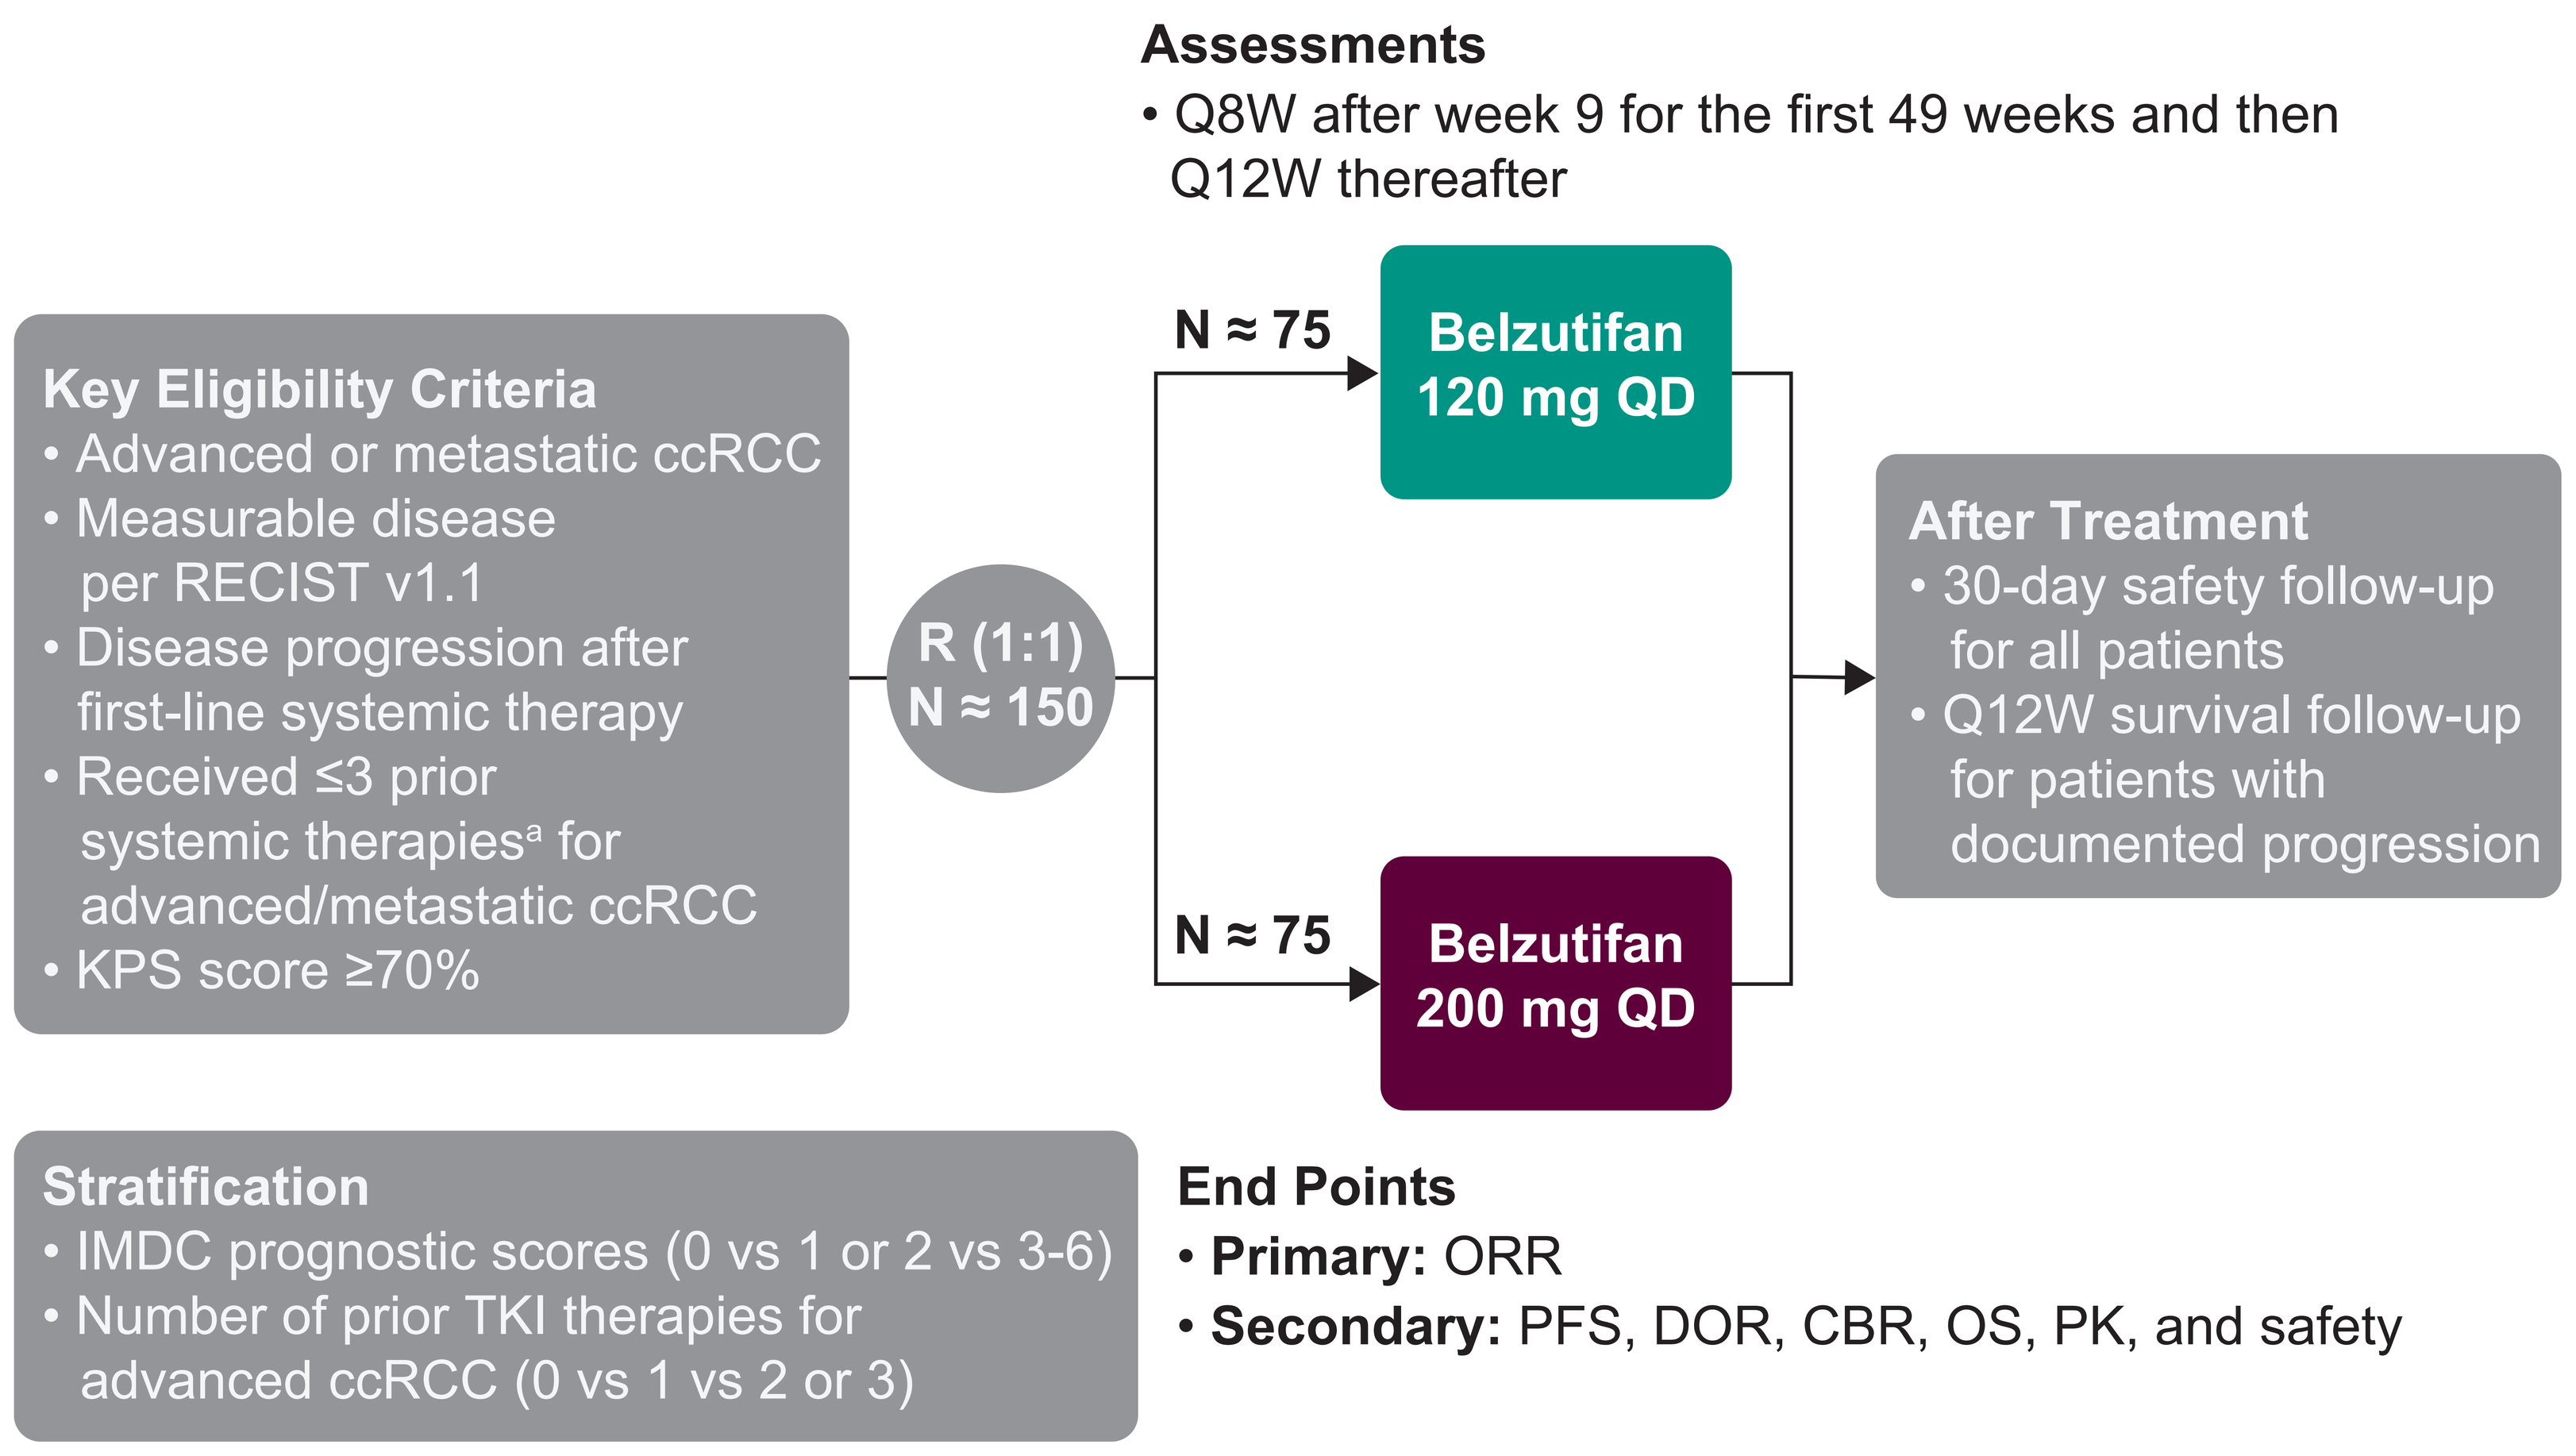
Assessments
• Q8W after week 9 for the first 49 weeks and then Q12W thereafter
Key Eligibility Criteria
• Advanced or metastatic ccRCC
• Measurable disease per RECIST v1.1
• Disease progression after first-line systemic therapy
• Received ≤3 prior systemic therapies[a] for advanced/metastatic ccRCC
• KPS score ≥70%
R (1:1)
N ≈ 150
N ≈ 75
Belzutifan 120 mg QD
N ≈ 75
Belzutifan 200 mg QD
After Treatment
• 30-day safety follow-up for all patients
• Q12W survival follow-up for patients with documented progression
Stratification
• IMDC prognostic scores (0 vs 1 or 2 vs 3-6)
• Number of prior TKI therapies for advanced ccRCC (0 vs 1 vs 2 or 3)
End Points
• Primary: ORR
• Secondary: PFS, DOR, CBR, OS, PK, and safety

KPS, Karnofsky Performance Status Scale; Q8W, every 8 weeks; Q12W, every 12 weeks; R, randomization.

[a]Prior systemic therapies must include first-line anti–PD-1/anti–PD-L1 + anti–CTLA-4 or anti–PD-1/anti–PD-L1 + VEGF-targeted TKI combination.

Patient Eligibility Criteria

Key Inclusion Criteria	Key Exclusion Criteria
• Age ≥18 years • Locally advanced/metastatic ccRCC (with or without sarcomatoid features) • Measurable disease per RECIST v1.1 by BICR • Disease progression after first-line systemic therapy with prior anti–PD-1/anti–PD-L1 + anti–CTLA-4 combination or anti–PD-1/anti–PD-L1 + VEGF-targeted TKI combination • Radiographic disease progression after the most recent regimen (if >1 prior regimen) • Has not previously received >3 systemic therapies for locally advanced or metastatic ccRCC • KPS score ≥70% assessed within 10 days before first dose of study drug	• Known central nervous system metastases and/or carcinomatous meningitis • Prior treatment with belzutifan or another HIF-2α inhibitor or any type of small molecule kinase inhibitor ≤2 weeks before randomization • Received any systemic anticancer antibody ≤4 weeks before randomization • Active infection necessitating systemic therapy • History of HIV infection • History of hepatitis B virus infection or active hepatitis C virus infection

Assessments and Follow-Up

- Radiologic evaluation will occur at week 9, then Q8W for 49 weeks, and Q12W thereafter
 - Patients who experience progression or begin a new anticancer regimen will enter the survival follow-up phase, in which they will be followed up for survival status approximately Q12W after the treatment discontinuation visit or discontinuation of follow-up imaging
- Adverse events (AEs) will be monitored throughout the study and graded in severity per Common Terminology Criteria for Adverse Events (CTCAE), version 5.0
 - AEs will be reported for 30 days after cessation of study drug
 - Serious AEs will be reported for 90 days after cessation of study drug or for 30 days if the patient begins a new anticancer therapy regimen
- A posttreatment safety follow-up visit will occur 30 days after the last dose of study drug is administered

Analysis

- The efficacy analysis will be performed for the intention-to-treat population (all randomly assigned patients)
 - The primary end point ORR will be evaluated using the stratified Miettinen and Nurminen method,[5] with strata weighted by sample size
 - The secondary end points PFS and OS will be summarized within each treatment group using the Kaplan-Meier method; the hazard ratio will be estimated using a stratified Cox regression model
- The safety analysis will be performed for the all-patients-as-treated population (all randomly assigned patients who received ≥1 dose of study drug)
 - For analyses in which 95% CIs will be provided for between-treatment differences in the percentage of participants who experience events, the Miettinen and Nurminen method will be used

Status

- Patients will be enrolled in the study in at least 9 countries (Australia, Belgium, Greece, Ireland, Israel, Netherlands, Russia, the United Kingdom, and the United States); patients are being recruited (**Figure 3**)

Figure 3. Countries With Sites of Enrollment (green)

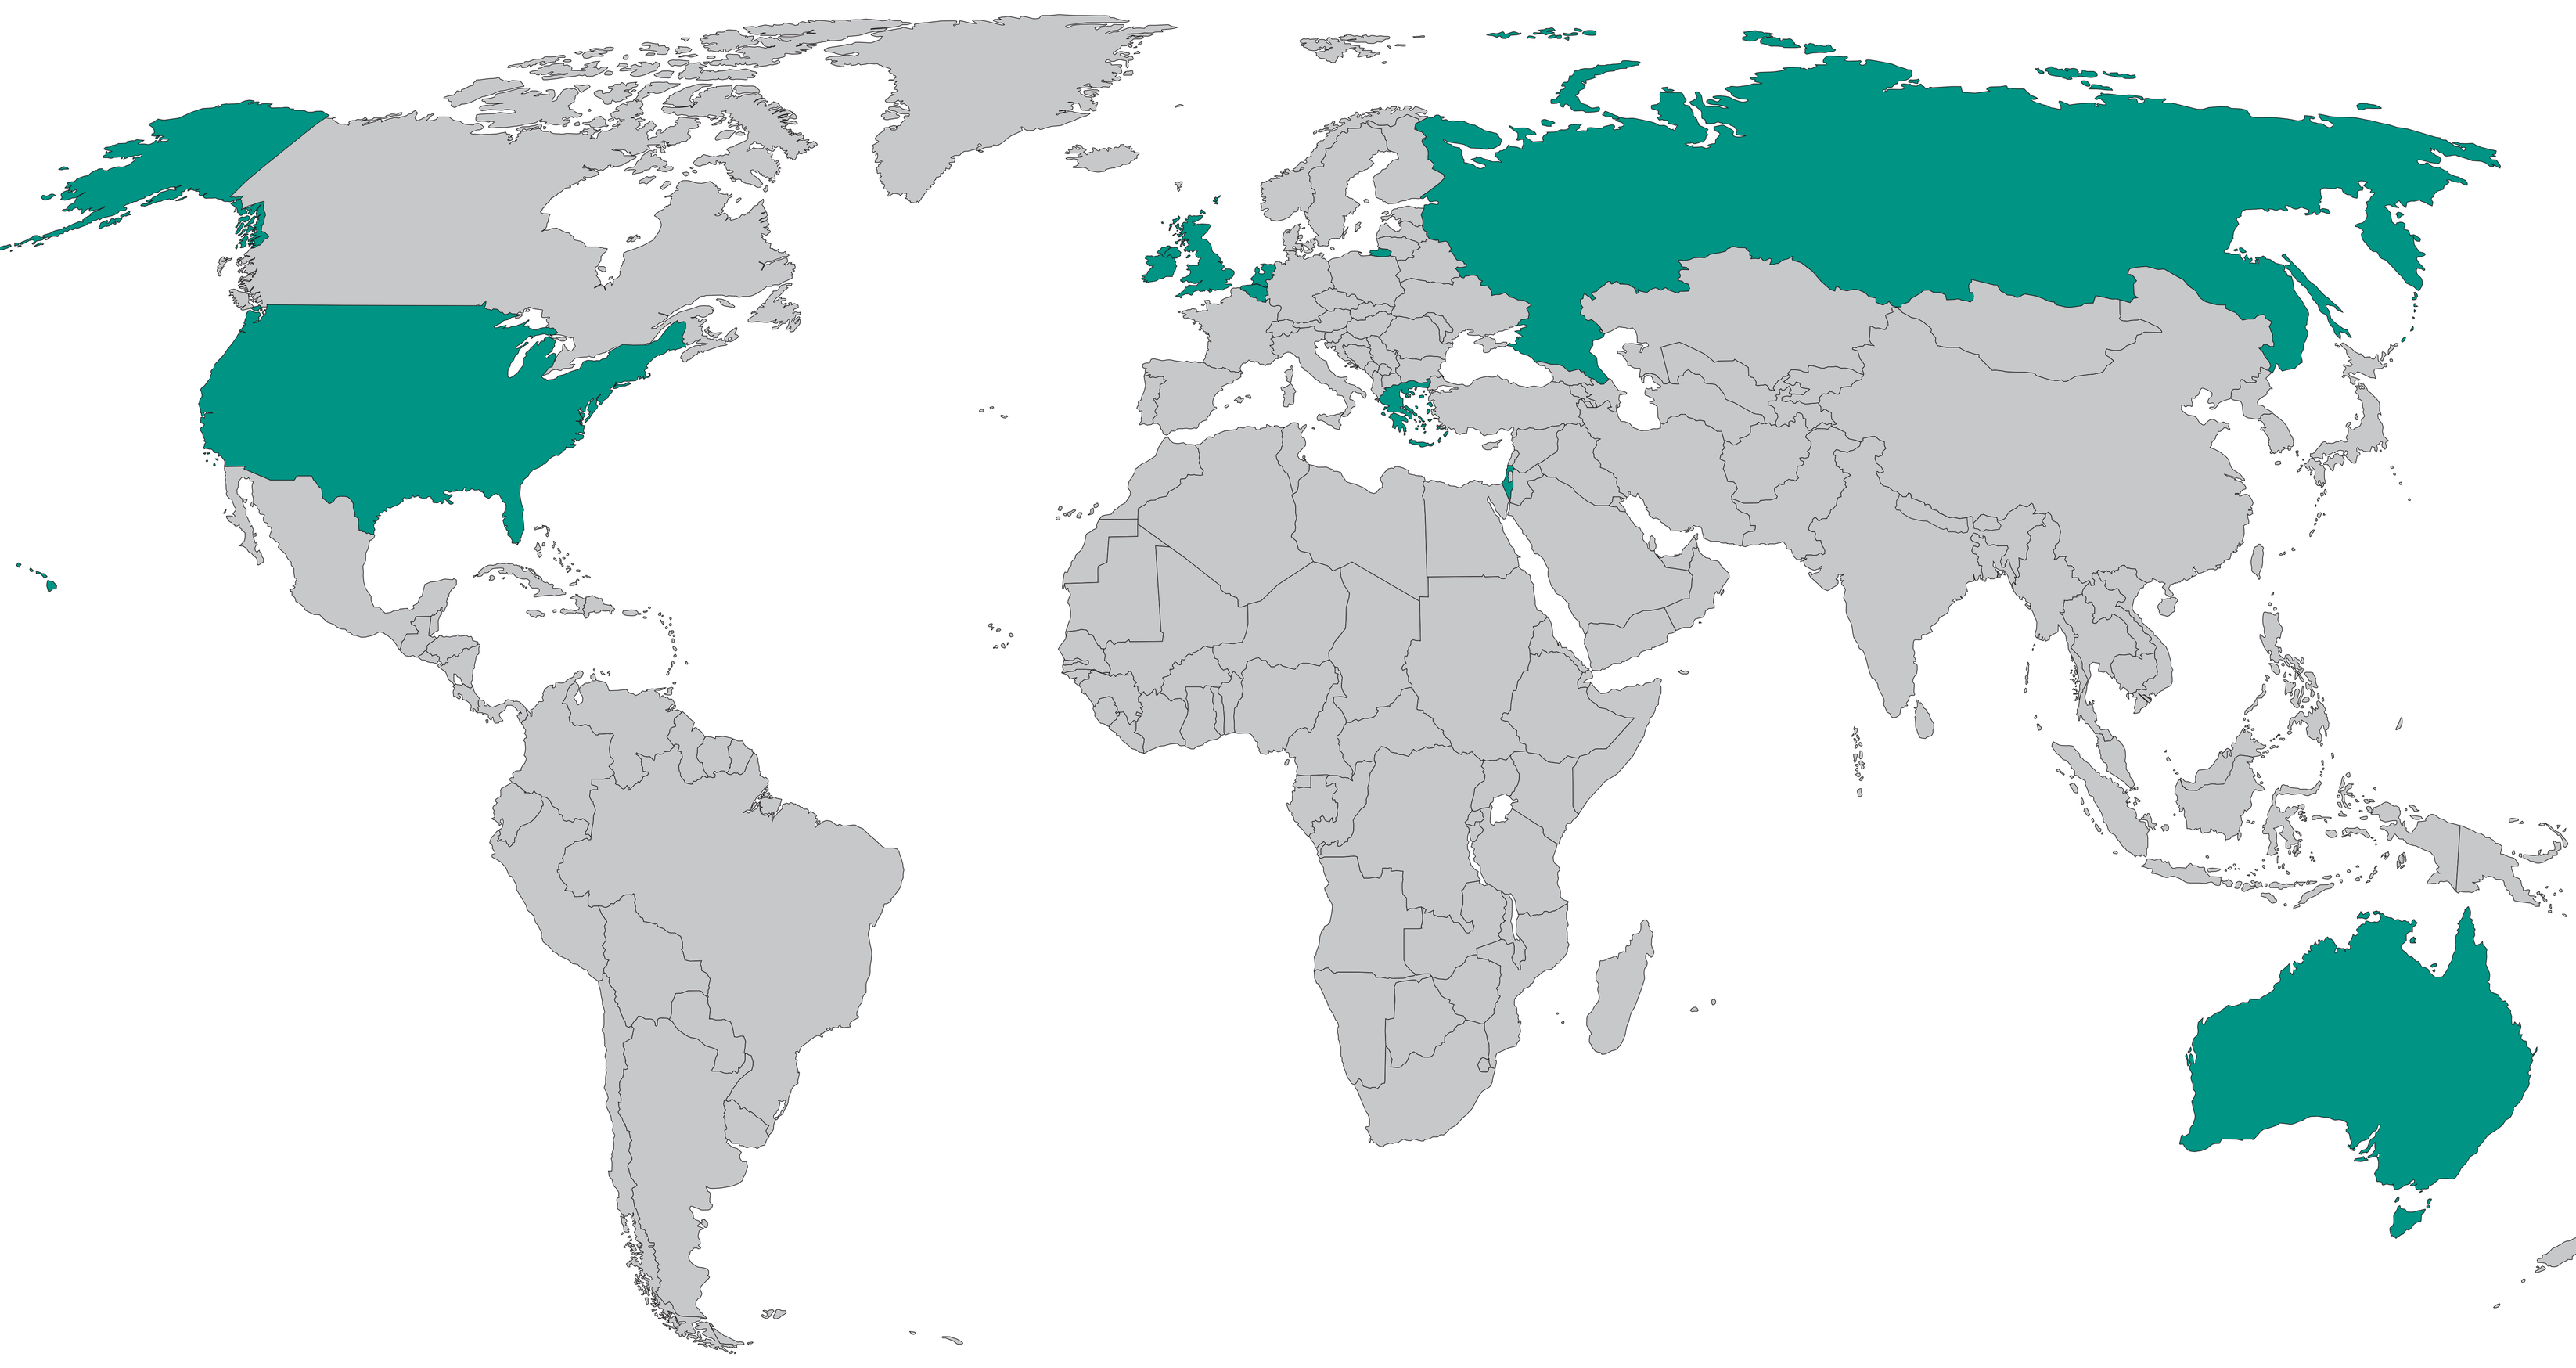

References

1. Linehan WM, Rickets CJ. *Nat Rev Urol*. 2019;16:539-552.
2. Wong SC et al. *Mol Cancer Ther*. 2018;17(suppl):B140.
3. Chouaib S et al. *Front Immunol*. 2012;3:21.
4. Bauer TM et al. *J Clin Oncol*. 2021;39(suppl 6). Abstract 273.
5. Miettinen O, Nurminen M. *Stat Med*. 1985;4:213-226.

Acknowledgments

The authors thank the patients and their families and investigators and site personnel. Medical writing and/or editorial assistance was provided by Shane Walton, PhD, and Matthew Grzywacz, PhD, of ApotheCom (Yardley, PA, USA). This assistance was funded by Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc., Kenilworth, NJ, USA. Funding for this research was provided by Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc., Kenilworth, NJ, USA.

Contact Information

Contact the author at mba41@georgetown.edu for questions or comments.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from IKCS and the author of this poster.

https://bit.ly/3vyloRh

Presented at the IKCS 2021: North America; November 5-6, 2021; Austin, Texas (Hybrid)

Copyright © 2021 Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc. All rights reserved.